Abstract 686

Mechanism of action of LAVA-051, a bispecific Vγ9Vδ2 T-cell engager, confirmed in the clinical setting

Roeland Lameris[1], Jurjen M. Ruben[2], Rob C. Roovers[2], Arnon P. Kater[3], Thilo Riedl[2], Victoria Iglesias[2], Annemiek Broijl[4], Ilse Tuinhof[2], Sanjana Umarale[2], Anton E.P. Adang[2], Tanja D. de Gruijl[1], Paul W.H.I. Parren[2], Benjamin Winograd[2], Hans J. van der Vliet[1,2]

LAVA THERAPEUTICS

INTRODUCTION

- LAVA-051: 27kD humanized bispecific single domain antibody (bsVHH) that directly engages CD1d and the Vδ2-TCR chain of Vγ9Vδ2-T cells to mediate potent killing of CD1d-expressing tumor cells.
- Engages 2 innate-like T cell populations with inherent antitumor activity: iNKT cells (type 1 NKT cells)(Nature Cancer 2020;1:1054-1065) and Vγ9Vδ2-T cells.

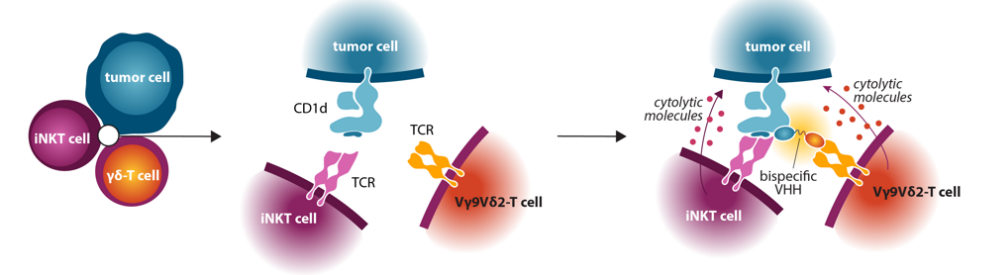

tumor cell
iNKT cell
γδ-T cell
CD1d
TCR
Vγ9Vδ2-T cell
cytolytic molecules
bispecific VHH

- CD1d: expressed by tumor cells in the majority of patients with CLL (chronic lymphocytic leukemia) and MM (multiple myeloma).
- This research investigates whether LAVA-051 pharmacodynamic data in the clinic are reflective of pre-clinical findings on the drug's mechanism of action. Data cut-off date 09-Sep-2022

SUMMARY AND CONCLUSION

- LAVA-051 binding to Vγ9Vδ2-T cells *in vitro* and *in vivo* lead to target-dependent Vγ9Vδ2-T cell engagement and activation (panels 1, 5, 8).
 - Reflected in phase 1 study and in NHP study (using a surrogate CD1d γδ T cell engager [TCE]) by a temporary decrease in circulating Vγ9Vδ2-T cells after dosing and concomitant upregulation of the activation markers CD25 and/or CD69.
 - Maximum measured Vγ9Vδ2-T cell receptor occupancy (RO) increased with higher dose cohorts in the clinic.
- LAVA-051 specifically induced Vγ9Vδ2-T and iNKT cell mediated killing of CLL and MM cells *in vitro*, while relatively sparing normal CD1d expressing cells (panel 2 and 3). In the clinic, first potential signs of activity towards tumor cells were observed. Also here, and in NHP study, normal CD1d expressing monocytes were not affected (panel 6 and 9).
- *In vitro* LAVA-051 mediated iNKT and Vγ9Vδ2-T activation is accompanied by induction of pro-inflammatory cytokines (panel 4). In NHP study and clinical setting, a mild and temporary increase in cytokines such as IL-6, IFN- and TNF was observed (panels 7 and 10). Importantly, there were no observed signs of cytokine release syndrome (CRS).
- In conclusion, LAVA-051 has demonstrated on-mechanism pharmacodynamics in the clinic to date, reflective of pre-clinical findings.
- Dose escalation continuing, including evaluation of subcutaneous dosing.

PRE-CLINICAL STUDIES

1 – DYNAMICS OF LAVA-051 BINDING AND Vγ9Vδ2-T/iNKT CELL ACTIVATION

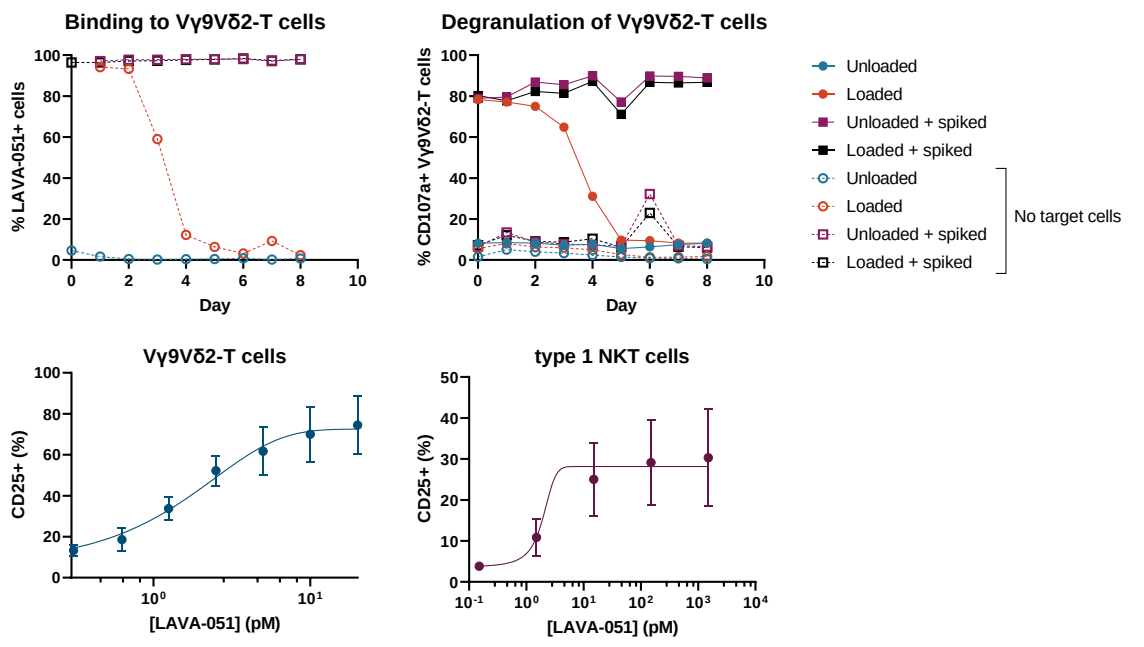

Binding to Vγ9Vδ2-T cells
% LAVA-051+ cells
Day
Degranulation of Vγ9Vδ2-T cells
% CD107a+ Vγ9Vδ2-T cells
Unloaded
Loaded
Unloaded + spiked
Loaded + spiked
No target cells
Vγ9Vδ2-T cells
CD25+ (%)
[LAVA-051] (pM)
type 1 NKT cells

2 - KILLING OF PATIENT-DERIVED CLL AND MM CELLS

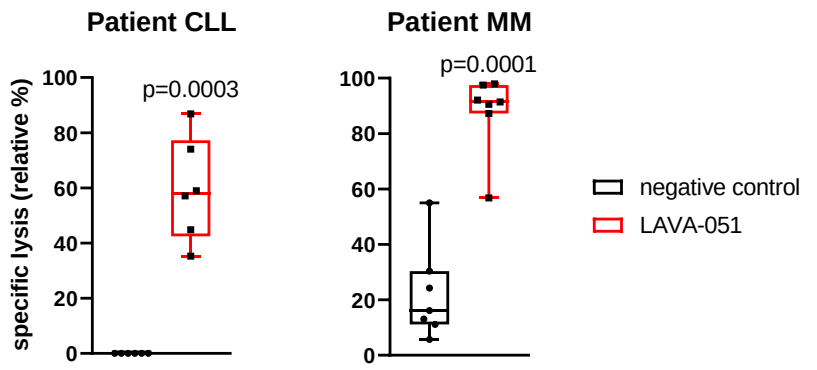

Patient CLL
Patient MM
specific lysis (relative %)
p=0.0003
p=0.0001
negative control
LAVA-051

3 - SPARING OF NORMAL CD1d EXPRESSING CELLS

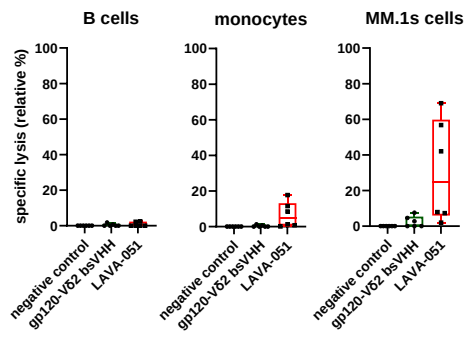

B cells
monocytes
MM.1s cells
specific lysis (relative %)
negative control
gp120-Vδ2 bsVHH
LAVA-051

4 – TRIGGERING OF iNKT AND Vγ9Vδ2-T CELL CYTOKINE PRODUCTION

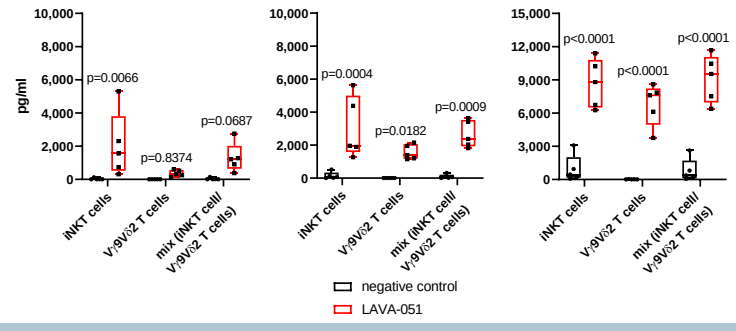

IL-2
TNF-α
IFN-γ
pg/ml
p=0.0066
p=0.8374
p=0.0687
p=0.0004
p=0.0182
p=0.0009
p<0.0001
iNKT cells
Vγ9Vδ2 T cells
mix (iNKT cell/ Vγ9Vδ2 T cells)
negative control
LAVA-051

NON-CLINICAL SAFETY STUDY

5 – DYNAMICS OF SURROGATE bsTCE BINDING AND Vγ9Vδ2-T ACTIVATION

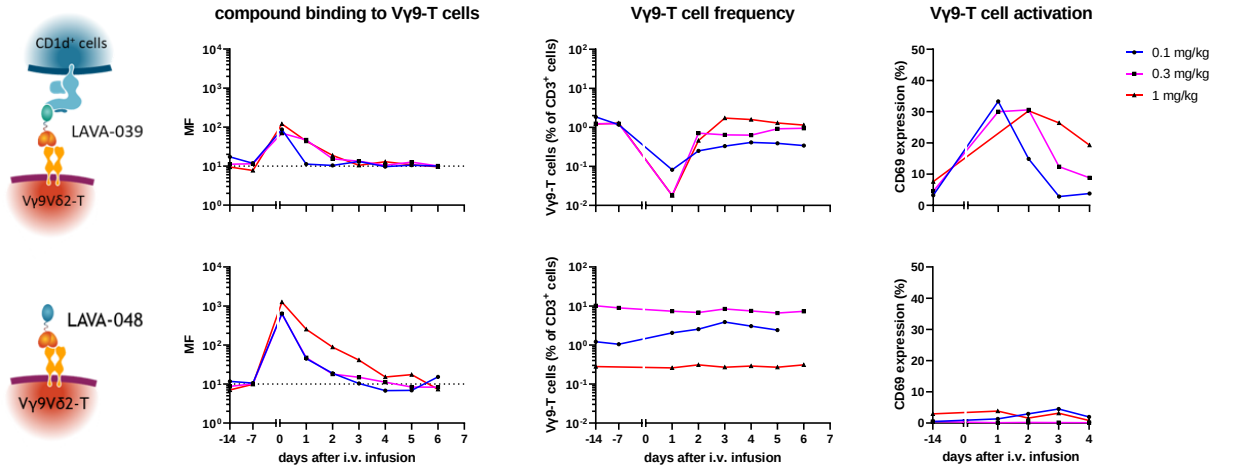

CD1d+ cells
LAVA-039
LAVA-048
Vγ9Vδ2-T
compound binding to Vγ9-T cells
Vγ9-T cell frequency
Vγ9-T cell activation
MF
Vγ9-T cells (% of CD3+ cells)
CD69 expression (%)
days after i.v. infusion
0.1 mg/kg
0.3 mg/kg
1 mg/kg

6 – NUMBERS OF CD1d EXPRESSING B CELLS AND MONOCYTES ARE NOT AFFECTED

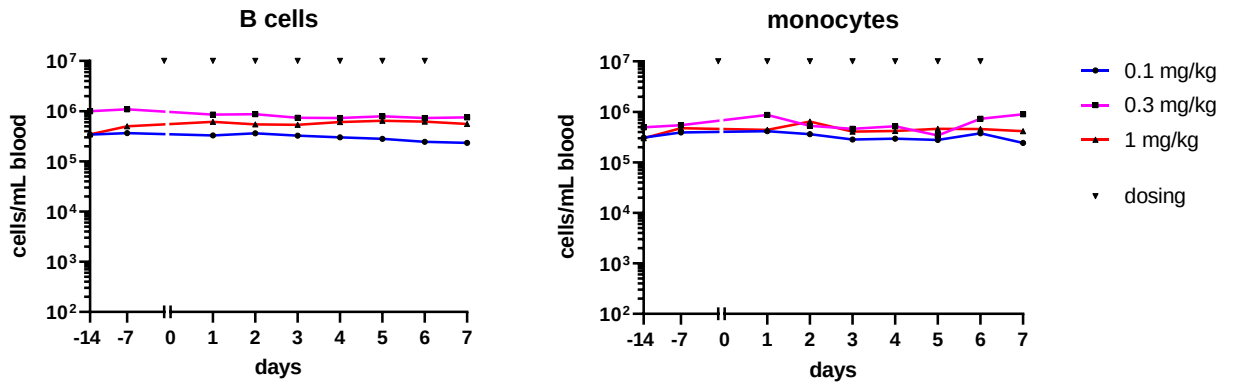

B cells
monocytes
cells/mL blood
days
0.1 mg/kg
0.3 mg/kg
1 mg/kg
dosing

7 – MILD AND TEMPORARY INCREASE IN CYTOKINES - NO SIGNS OF CRS

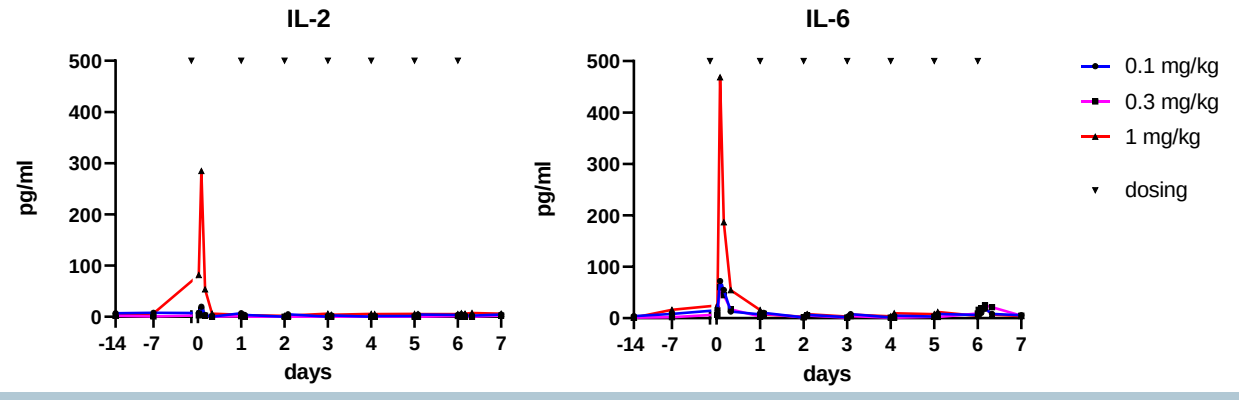

IL-2
IL-6
pg/ml
days
0.1 mg/kg
0.3 mg/kg
1 mg/kg
dosing

METHODS

1. TOP: Left: Vγ9Vδ2-T cells were incubated with 100 nM LAVA-051, washed, and binding of LAVA-051 was detected by flow cytometry. LAVA-051 'loaded' Vγ9Vδ2-T cells were placed in culture, washed every day, analyzed for binding of LAVA-051 and re-cultured. Right: In parallel, the LAVA-051 'loaded' Vγ9Vδ2-T cells were tested in a 4 hour activation (degranulation) test using CCRF-CEM tumor cells as target cells.
 BOTTOM: O/N culture Vγ9Vδ2-T cells/iNKT cells and MM1s.CD1d cells ± LAVA-051 (E:T ratio of 1:1). CD25 expression determined by flow cytometry.
2. Cytotoxicity towards patient CLL and MM cells after 16 hr coculture of iNKT and Vγ9Vδ2-T cells (1:1 mix) and patient PBMC or BMMC (E:T=1:2) ± 50nM LAVA-051.
3. Lysis after O/N culture of PBMC and CD1d+ MM.1s cells (ratio 10:1) ± 50 nM gp120-Vδ2 bsVHH or LAVA-051.
4. Cytokine secretion by iNKT, Vγ9Vδ2-T cells or a mixture (ratio 1:1) after a 24 h culture with CD1d+ MM.1s cells ± LAVA-051(50 nM), measured by cytometric bead array.

5. – 7. Non-clinical safety study in cynomolgus monkey: Surrogate bs-γδTCEs LAVA-039 (fully cross-reactive) and LAVA-048 (Vγ9 cross-reactive) were generated because of lack of LAVA-051 NHP cross-reactivity. Part I: single dose: 0.1, 0.3 or 1 mg/kg (i.v.; n=1/dose). Part II: 7 daily doses: same dose range (i.v.; n=1/dose). bs-γδTCE binding, frequency and activation status of Vγ9Vδ2-T and other immune cells (shown for LAVA-039 only) determined by flow cytometry. Cytokine levels (shown for LAVA-039 only) determined by cytometric bead array.

8. – 10. Panel to determine specific pharmacodynamic parameters in circulation included in ongoing phase 1 study of LAVA-051 in patients with relapsed/refractory MM or CLL (NCT04887259): Vγ9Vδ2-TCR occupancy (RO) with LAVA-051; frequency and activation status of Vγ9Vδ2-T and other immune cells; cytokine levels.

FIH CLINICAL TRIAL

8 – DYNAMICS OF LAVA-051 BINDING AND Vγ9Vδ2-T ACTIVATION

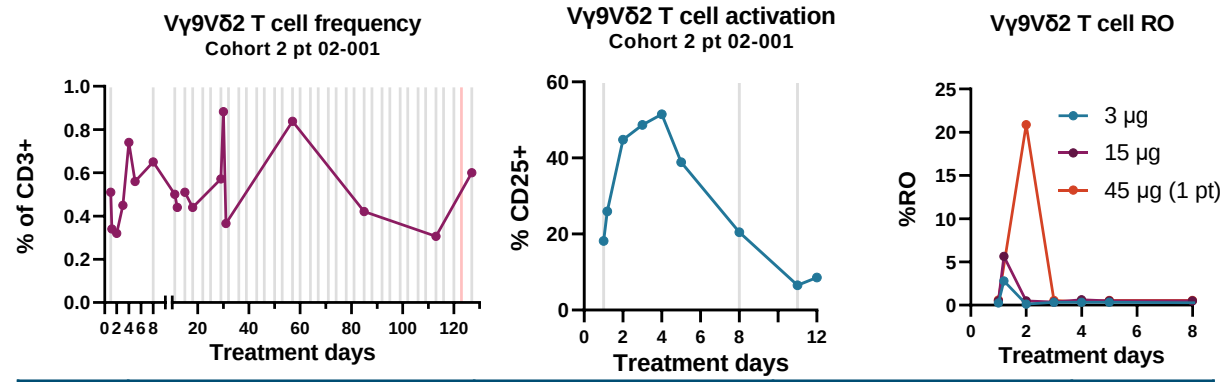

Vγ9Vδ2 T cell frequency
Cohort 2 pt 02-001
% of CD3+
Vγ9Vδ2 T cell activation
Cohort 2 pt 02-001
% CD25+
Vγ9Vδ2 T cell RO
%RO
3 μg
15 μg
45 μg (1 pt)
Treatment days

Dose (μg)	Vγ9Vδ2 T cell frequency (% of CD3+)		% CD25+		% CD69+		RO_{max} (%)
	Baseline	Post-dosing (range)	Baseline	Post-dosing (max)	Baseline	Post-dosing (max)	
0.45	0.00	ND	ND	ND	ND	ND	NC
3	0.51	0.31 - 0.74	18.2	51.5	48.5	88.9	2.8
15	1.46	0.02 - 1.08	0.9	7.9	13.3	83.3	5.6
45	0.41	0 - 0.42	15.2	35.6	30.3	86.7	20.9
45	0.45	0 - 0.10	12.5	55.6	25	0	NC
45	0.05	0 - 0.07	0	50.0	0	100	0.6*

ND = not detectable; NC = RO_{max} could not be calculated; * RO_{max} could only be determined at 72 hr after end of infusion

9 – POTENTIAL SIGNS OF ACTIVITY WHILE SPARING NORMAL CD1d EXPRESSING CELLS

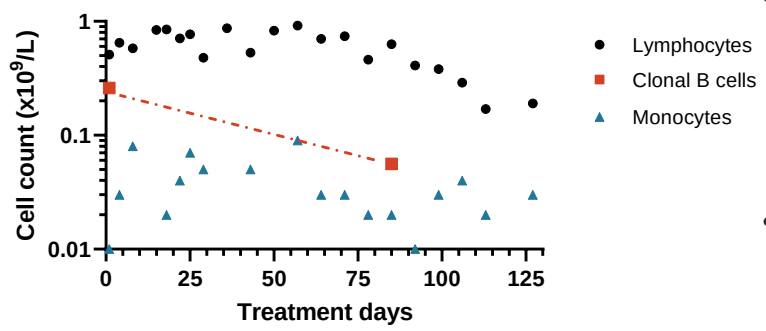

Cell count (x10^9/L)
Treatment days
Lymphocytes
Clonal B cells
Monocytes

- Percent of PB clonal B cells of R/R CLL patient (15 μg) decreased from 41.8% at baseline to 8.9% at start of Cycle 4.
- Numbers of normal CD1d expressing monocytes remained stable over time.

10 – MILD AND TEMPORARY INCREASE IN CYTOKINES - NO SIGNS OF CRS

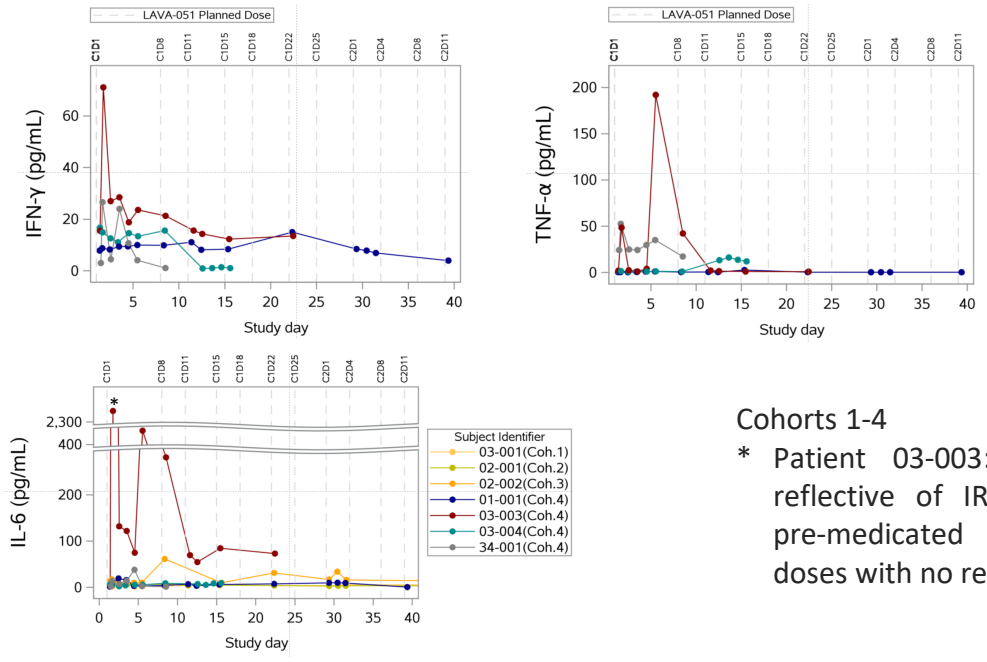

LAVA-051 Planned Dose
IFN-γ (pg/mL)
TNF-α (pg/mL)
IL-6 (pg/mL)
Study day
Subject Identifier
03-001(Coh.1)
02-001(Coh.2)
02-002(Coh.3)
01-001(Coh.4)
03-003(Coh.4)
03-004(Coh.4)
34-001(Coh.4)

Cohort 4 (45 μg)

Cohorts 1-4

* Patient 03-003: IL-6 increase reflective of IRR; patient was pre-medicated for subsequent doses with no recurrence of IRR

[1]Department of Medical Oncology, Cancer Center Amsterdam, Amsterdam UMC, Amsterdam, Netherlands, [2]LAVA Therapeutics N.V., Utrecht, Netherlands, [3]Department of Hematology, Cancer Center Amsterdam, Amsterdam UMC, Amsterdam, Netherlands, [4]Department of Hematology, Erasmus MC, Rotterdam, Netherlands
Contact: b.winograd@lavatherapeutics.com